Prevalence of Prostate Cancer Clinical States in US: Estimates Using a Dynamic Progression Model

K. Solo,[1] M. Mehra,[2] R. Dhawan,[2] J. Valant,[1] and H.I. Scher[3]

[1]Lexidyne, LLC, Colorado Springs, CO, USA; [2]Janssen Global Services, LLC, Raritan, NJ, USA; [3]Memorial Sloan-Kettering Cancer Center, New York, NY, USA

INTRODUCTION

- Prostate cancer is a significant cause of morbidity and mortality in the US, with an age-adjusted incidence of 156.0 cases per 100,000 men per year. The estimated incidence in 2010 was 217,730 new cases, with 32,050 deaths.[1]
- Despite the high overall incidence of prostate cancer, most newly diagnosed patients in the US present with clinically localized disease[1] (Figure 1), many of which are indolent cancers that, left untreated, would not result in symptoms, metastases, or death from disease.[2]
- New methods are urgently required to identify those tumors that require treatment based on the probability (risk) of clinically significant disease progression and death from disease. For these men, more effective, integrated, multimodality treatment approaches that include systemic therapies are needed.
- Cancer is a dynamic disease that can change as it evolves/progresses. This is due to the intrinsic properties of the tumor itself, as well as the specific therapies to which it has been exposed.
- Understanding the prognosis, impact of available treatments, and probability of progression at all points of the disease continuum is therefore essential to guide management decisions and improve patient outcomes. These issues are not addressed by traditional staging systems or nomograms, which typically only consider the disease at the time of diagnosis.[3]
 - Dynamic disease progression models that consider both the untreated natural and post-treatment history of a disease divide the disease continuum from diagnosis to death into distinct clinical states. The states represent key points in the illness where therapeutic decisions need to be made.[4]
- Here we present such a model for prostate cancer, based on US epidemiological estimates of disease incidence, progression, and mortality.
 - This multistate model, derived from the stocks and flows concept, allows the estimation of the annual prevalence ("stocks") of each clinical state, as well as of annual progression ("flows") between clinical states.
 - Based on the currently available treatments for patients in each clinical state and the disease prevalence at each state, the model simulates all-cause mortality at those states, as well as the unidirectional movement of patients from one clinical state to the next.
 - The model also provides a flexible structure to simulate the impact of novel prostate cancer therapies on patient progression and survival and, thus, guide patient management.

Figure 1. Incidence of Prostate Cancer in the US Between 1990 and 2009, by Stage of Disease at Diagnosis[1]

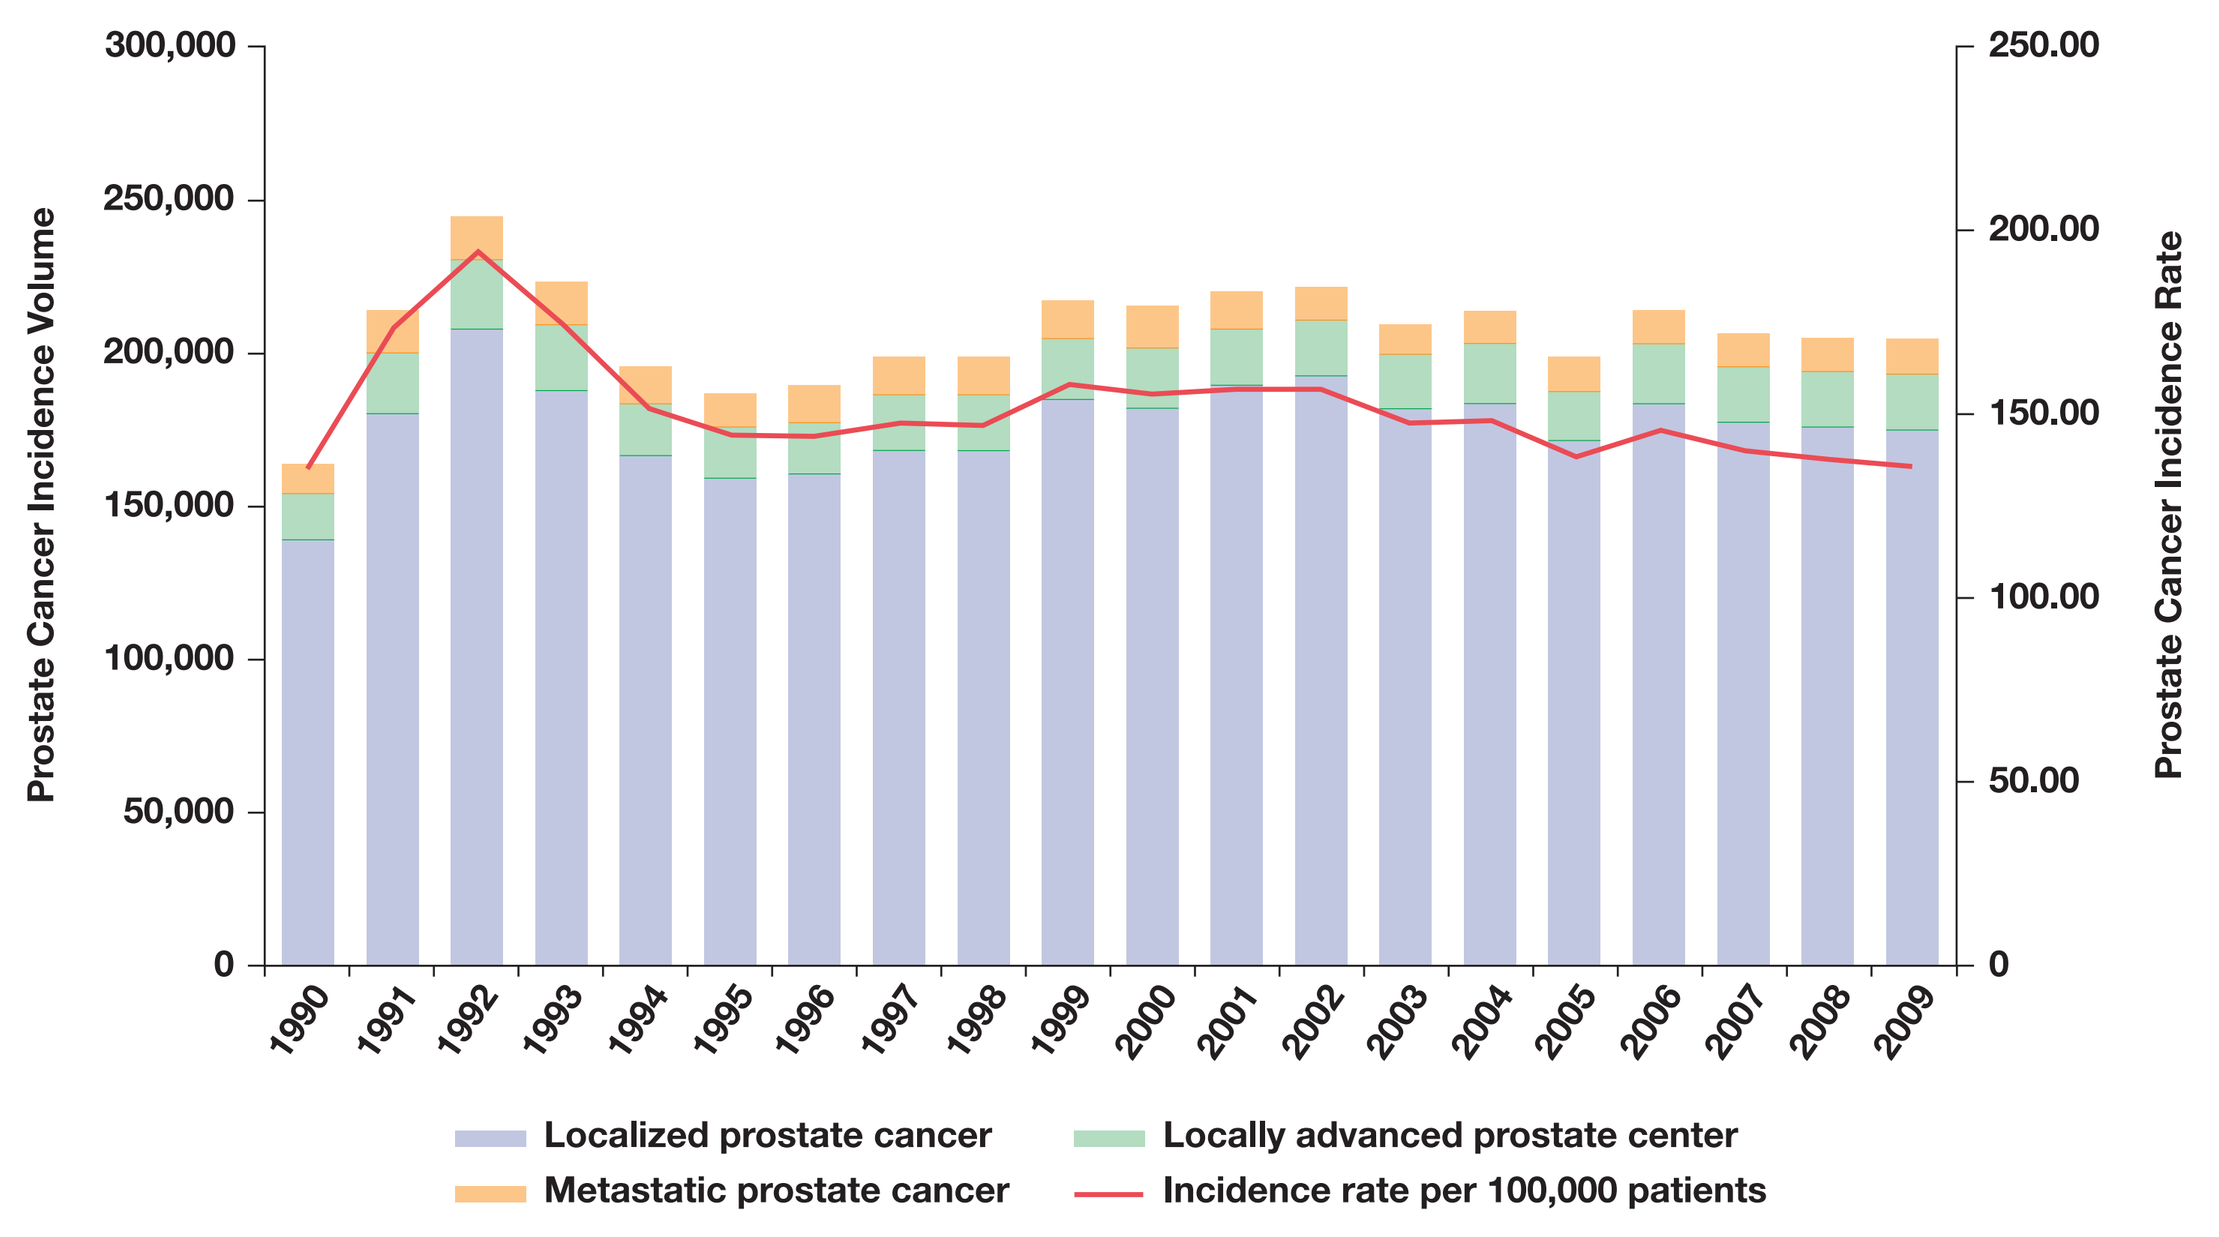
300,000
250,000
200,000
150,000
100,000
50,000
0
Prostate Cancer Incidence Volume
250.00
200.00
150.00
100.00
50.00
0
Prostate Cancer Incidence Rate
1990
1991
1992
1993
1994
1995
1996
1997
1998
1999
2000
2001
2002
2003
2004
2005
2006
2007
2008
2009
Localized prostate cancer
Locally advanced prostate center
Metastatic prostate cancer
Incidence rate per 100,000 patients

METHODS

- An intervention-based simulation methodology was developed. The simulation modeled the progression of prostate cancer patients through a series of 8 Markov states, which are defined in Table 1.
 - Each of these represents a distinct clinical state of the disease, defined by the status of the primary tumor, presence/absence of clinically detectable metastatic disease, prior treatment, and serum testosterone levels (noncastrate/castrate).[4]
 - These states are easily recognized, reproducible, and form the basis for clinical investigation of therapeutic interventions in prostate cancer, including castration-resistant disease (CRPC), the lethal variant of the illness.
- This was a dynamic model (see Figure 2 for a graphic representation of the model structure). Patients could enter a specific state ("inflow") as the result of either a) newly diagnosed cancer, b) recurrence, or c) progression. The model also accounts for all-cause mortality ("outflow") associated with each state over time.
 - Untreated, newly diagnosed patients with prostate cancer entered ("flowed") into these clinical states: active surveillance (localized prostate cancer not undergoing therapy), L_1 (localized prostate cancer treated with definitive local therapy), and M_1 (newly diagnosed metastatic, non-CRPC). Newly diagnosed patients could also flow into state L_2 (nonmetastatic, non-CRPC treated with androgen deprivation therapy). Patients also flowed into L_2 as a result of disease progression from active surveillance or L_1. Inflows into clinical states R (nonmetastatic CRPC), M_2 (asymptomatic metastatic CRPC), M_3 (symptomatic metastatic CRPC undergoing first-line chemotherapy), and M_4 (symptomatic metastatic CRPC after failed first-line chemotherapy) were only the result of disease progression (Figure 2).
 - In this model, patients could only move forward from state to state, and could not move backward into a previous state.
 - Patient flow between clinical states, as well as all-cause mortality at each state, were estimated using hazard rates derived from previously published Kaplan-Meier progression survival and overall survival curves specific to each clinical state (Table 1).[5-18] Trials reported after December 2009 are not included in the current version of the model, but will be incorporated into planned updates.
- The model was based on a 19-year simulation initiating in 1990, using annual prostate cancer incidence and distribution across the 4 clinical states at diagnosis (active surveillance, L_1, L_2, and M_1).
 - For each subsequent year until 2009, additional incidence cohorts were added into the model.
 - All annual incidence rates were US estimates obtained from the Surveillance, Epidemiology and End Results (SEER) registry.[1]
- The model estimated prevalent patients within each clinical state at a specific point in time. The net number of prevalent patients included those with active disease (ie, receiving treatment for prostate cancer) and those in remission (ie, not on treatment and did not progress or die).
- The model, as described above, was custom programmed in the Java™ programming language.

Table 1. Definitions of Clinical States (and Data Sources Used to Determine Annual Hazard Rates for Progression and Mortality) That Formed Part of This Disease Progression Model

Clinical state	Definition → currently recommended treatment	Relevant data sources
Active Surveillance	Localized prostate cancer → no treatment	Shappley, et al. 2009 (342 pts who deferred treatment > 1 yr after initial diagnosis)
L_1	Localized prostate cancer → definitive local therapy	Moreira, et al. 2009 (1,156 pts who underwent radical prostatectomy, excluding pts treated with preoperative hormonal therapy or radiotherapy) Merglen, et al. 2007 (844 pts with diagnosis of localized [stage T1, T2, T3, and M0] PC) Bolla, et al. 2005 (1,005 pts with previously untreated, histologically proven prostate adenocarcinoma [stage T0–3, nodal stage N0, no distant metastases, and pathologic stage pT2–3 N0 with ≥ 1 of risk factors: capsule perforation, positive surgical margins, invasion of seminal vesicles])
L_2	Nonmetastatic non-CRPC → ADT (Note: Includes pts who received ADT following definitive local therapy, who will be treated as a separate clinical state in an updated model)	Antonarakis, et al. 2007 (Pts with localized or locally advanced PC receiving immediate hormone therapy as adjunct to radiation therapy [n = 5,987], radical prostatectomy [n = 1,885], or standalone therapy [n = 3,233]) Pilepich, et al. 2001 (471 PC pts with bulky primary tumors, clinical stage T2–T4) Ross, et al. 2008 (553 PC pts treated with ADT [orchiectomy or LHRH], with or without an antiandrogen for nonadjuvant hormone-sensitive PC)
R	Nonmetastatic CRPC postsurgery/radiation/ADT → continue ADT	Nelson, et al. 2008 (941 pts with histologically confirmed, nonmetastatic hormone-resistant PC; had undergone surgical or pharmacologic castration at least 3 months before randomization; and had castrate testosterone levels at screening) Madan, et al. 2008 (42 pts with nonmetastatic CRPC determined by increasing serum PSA despite castrate levels of testosterone [< 50 ng/dL] with no radiographic evidence of metastatic disease)
M_1	Metastatic non-CRPC (newly diagnosed metastatic prostate cancer) → ADT	Chouieri, et al. 2008 (179 PC pts treated with ADT for metastatic HSPC with ≥ 2 serum PSA determinations before PSA nadir and ≥ 1 serum PSA value available within 1 month of ADT initiation)
M_2	Asymptomatic mCRPC → 1st-line chemotherapy[b]	Oudard, et al. 2009 (145 pts who had progressed on androgen-blockade therapy, and after withdrawal of antiandrogen therapy had an ECOG-PS ≤ 2 and a castrate serum testosterone level < 50 ng/mL)
M_3	Symptomatic mCRPC → 1st-line chemotherapy	Petrylak, et al. 2004 (770 pts with advanced refractory PC [pathologically confirmed adenocarcinoma of prostate and progressive metastatic disease, stage D1 or D2] despite androgen ablative therapy and cessation of antiandrogen treatment) Tannock, et al. 2004 (1,006 pts with metastatic hormone-resistant PC [histologically/cytologically confirmed adenocarcinoma of prostate with clinical or radiologic evidence of metastatic disease, had had disease progression during hormonal therapy, and were receiving primary androgen-ablation therapy as maintenance therapy])
M_4	Symptomatic mCRPC after failed 1st-line chemotherapy → 2nd-line chemotherapy[c]	Sternberg, et al. 2009 (950 pts with stage D2 [TxNxM1] metastatic adenocarcinoma of prostate and disease progression after ≥ 2 cycles of 1 prior chemotherapy regimen)

[a]Between 1990 and 2009 in the US, according to data from the SEER registry. [b]Sipuleucel-T is approved for this patient population, but relevant clinical trials were not included for purposes of estimating hazard ratios. [c]Both abiraterone acetate plus prednisone and cabazitaxel plus prednisone are approved for this patient population, but relevant clinical trials were not included for purposes of estimating hazard ratios.
ADT, androgen deprivation therapy (hormone therapy); ECOG-PS, Eastern Cooperative Oncology Group-performance status; HSPC, hormone-sensitive prostate cancer.

Figure 2. Patient Flow Between the Distinct Prostate Cancer Disease States in 2009 According to the Disease Progression Model

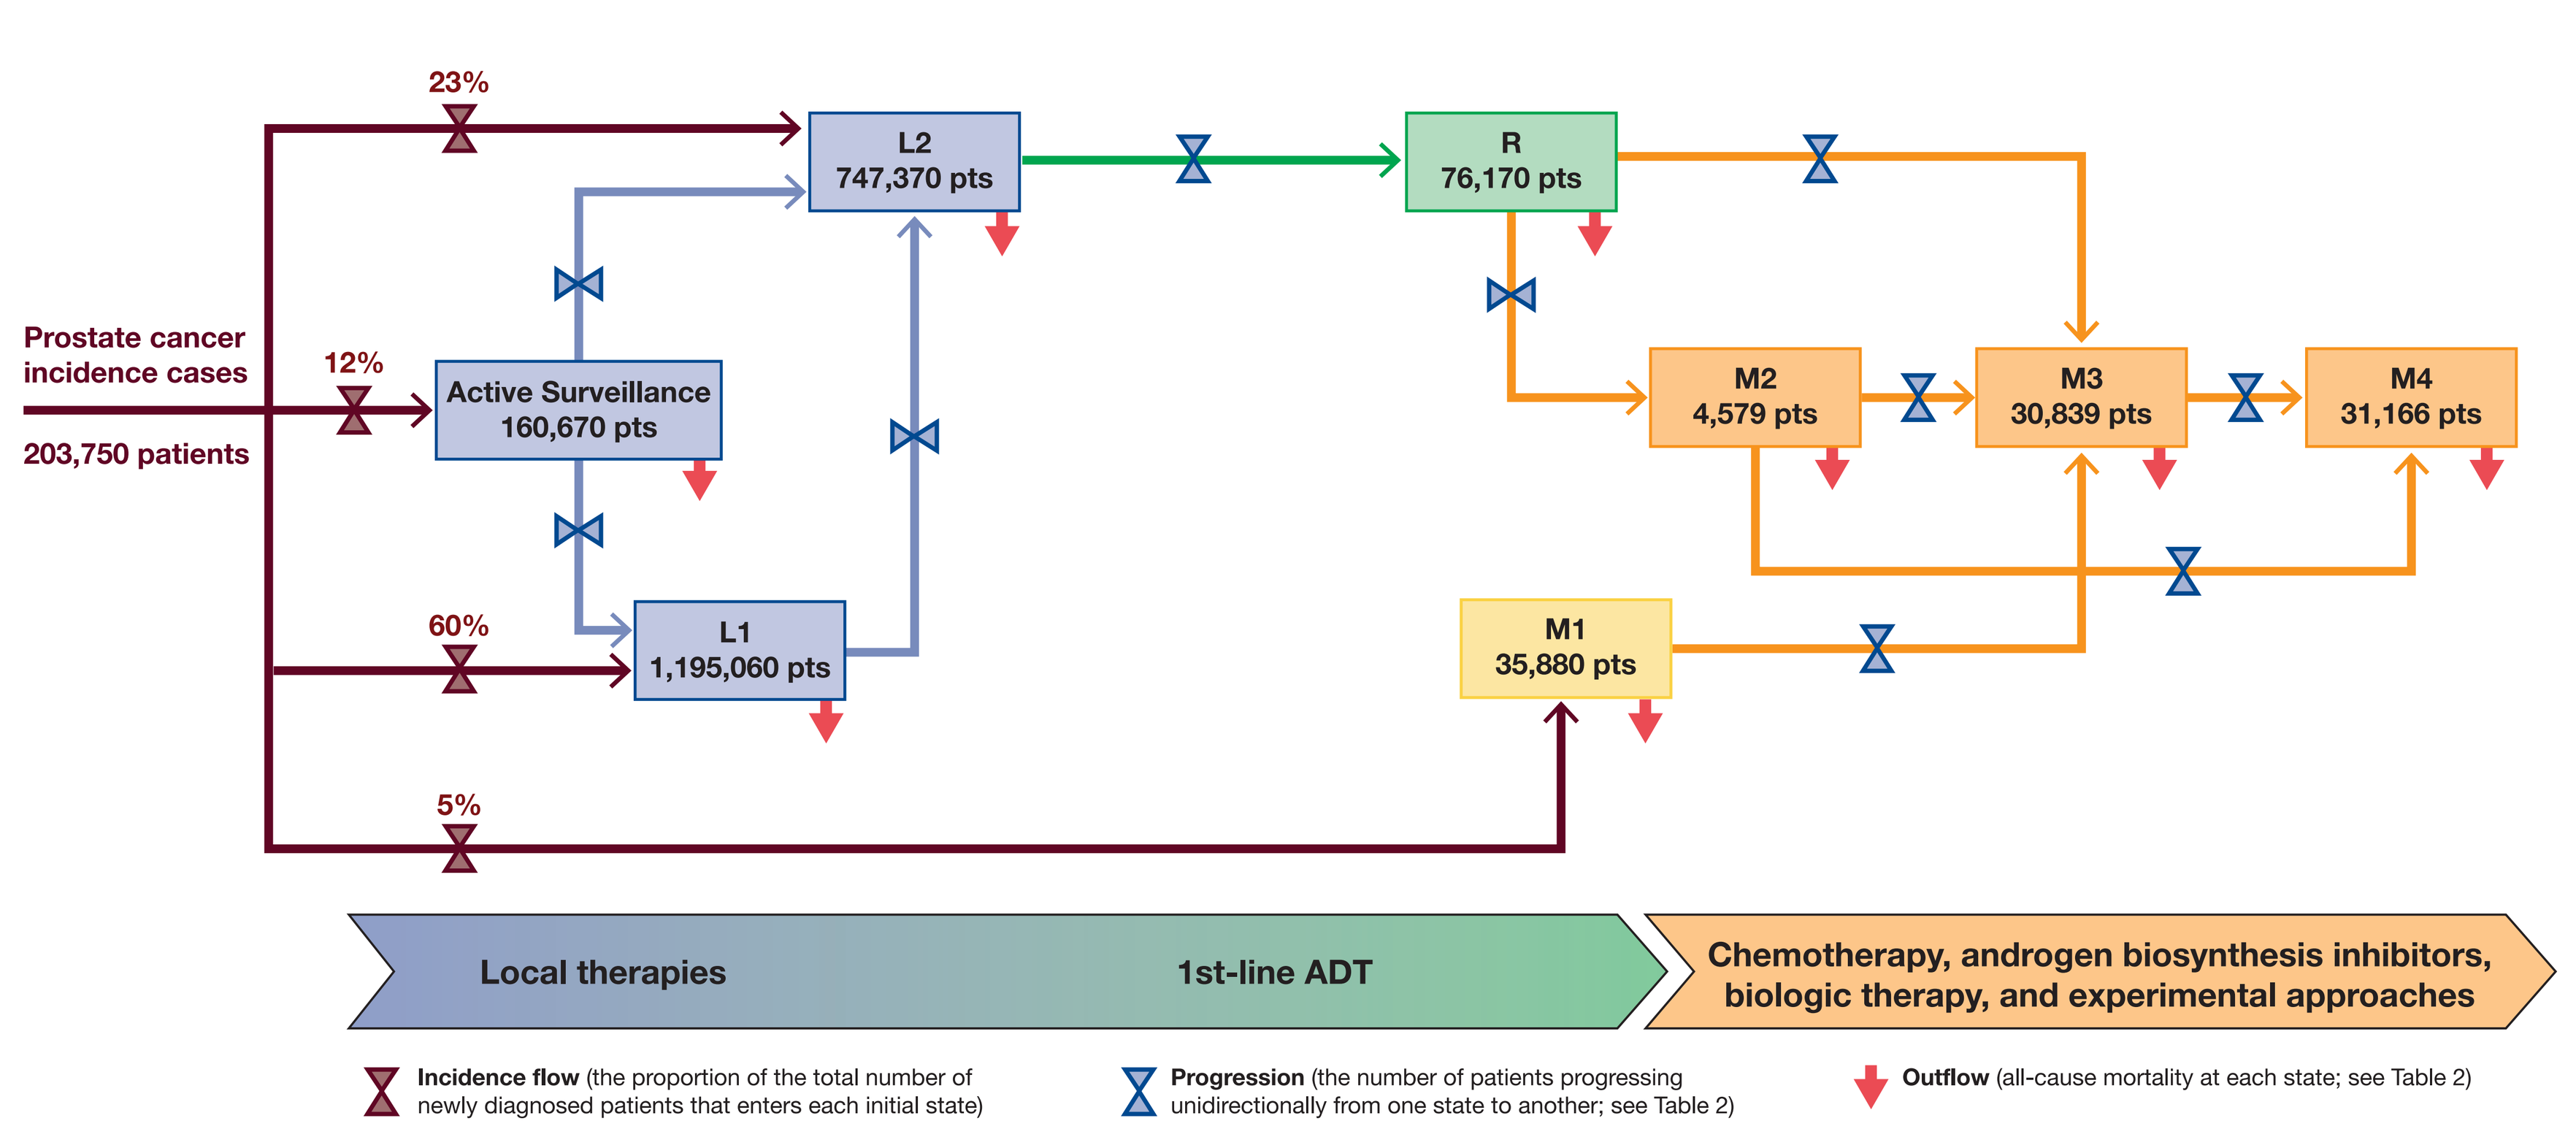
Prostate cancer incidence cases
203,750 patients
23%
12%
60%
5%
L2
747,370 pts
R
76,170 pts
Active Surveillance
160,670 pts
L1
1,195,060 pts
M2
4,579 pts
M3
30,839 pts
M4
31,166 pts
M1
35,880 pts
Local therapies
1st-line ADT
Chemotherapy, androgen biosynthesis inhibitors, biologic therapy, and experimental approaches
Incidence flow (the proportion of the total number of newly diagnosed patients that enters each initial state)
Progression (the number of patients progressing unidirectionally from one state to another; see Table 2)
Outflow (all-cause mortality at each state; see Table 2)

RESULTS

- Prevalent patients in each clinical state, incidence flow, and patient flows between the distinct prostate cancer disease states for 2009 are shown in Figure 2.
- Based on the 19-year simulation, the model estimated a prostate cancer prevalence of 2.3 million patients in the US for 2009, which aligns with a previously reported estimate of more than 2.1 million.[1]
 - Furthermore, the simulation indicated that 95.5% (n = 2,179,270) of prevalent prostate cancer patients in the US would have localized or locally advanced disease (active surveillance, L_1-L_2, R) and 4.5% (n = 102,464) would have metastatic disease (M_1-M_4; Table 2).
 - The numbers of patients progressing from and dying at each clinical state in 2009 are also shown in Table 2.
 - Over the course of 2009, the model estimated an incident flow of about 31,000 mCRPC cases, with 27,090 progressing from segment R and 3,797 from segment M_1 (Figure 3).
- The model estimated approximately 161,700 deaths (all-cause mortality) among US prostate cancer patients in 2009.
 - Of these overall deaths, about 30,100 (19%) were estimated to occur in patients with mCRPC, in whom mortality can be primarily attributed to prostate cancer. This number aligns with the 27,360 prostate cancer deaths in 2009 estimated by the American Cancer Society, based on US mortality data from 1969 to 2006.[19]

Table 2. Prevalence of Prostate Cancer and Active Prostate Cancer in 2009 According to the Disease Progression Model

Clinical state	2009 Prevalence, n	Active disease[a], n (%[b])	Outflow—progression n (%[b])	Outflow—mortality n (%[b])
Active Surveillance	160,670	–	16,220 (10.0)	14,540 (9.0)
L_1	1,195,060	83,380 (7.0)	61,070 (5.1)	44,750 (3.7)
L_2	747,370	142,840 (19.1)	41,570 (5.6)	54,470 (7.3)
R	76,170	48,450 (63.6)	27,090 (35.6)	10,630 (14.0)
M_1	35,880	17,493 (48.8)	3,797 (10.6)	7,202 (20.1)
M_2	4,579	3,712 (81.1)	2,307 (50.4)	1,682 (36.7)
M_3	30,839	26,928 (87.3)	17,500 (56.7)	10,476 (34.0)
M_4	31,166	27,406 (88.0)	–	17,973 (57.7)
Total prevalence	**2,281,734**	**350,209**	**–**	**–**

[a]Patients who are receiving prostate cancer treatment and are not in remission. [b]All proportions are based on the overall 2009 prevalence of each state.

Figure 3. Patient Flow Through Nonmetastatic and Metastatic Prostate Cancer Disease States in 2009 According to the Disease Progression Model

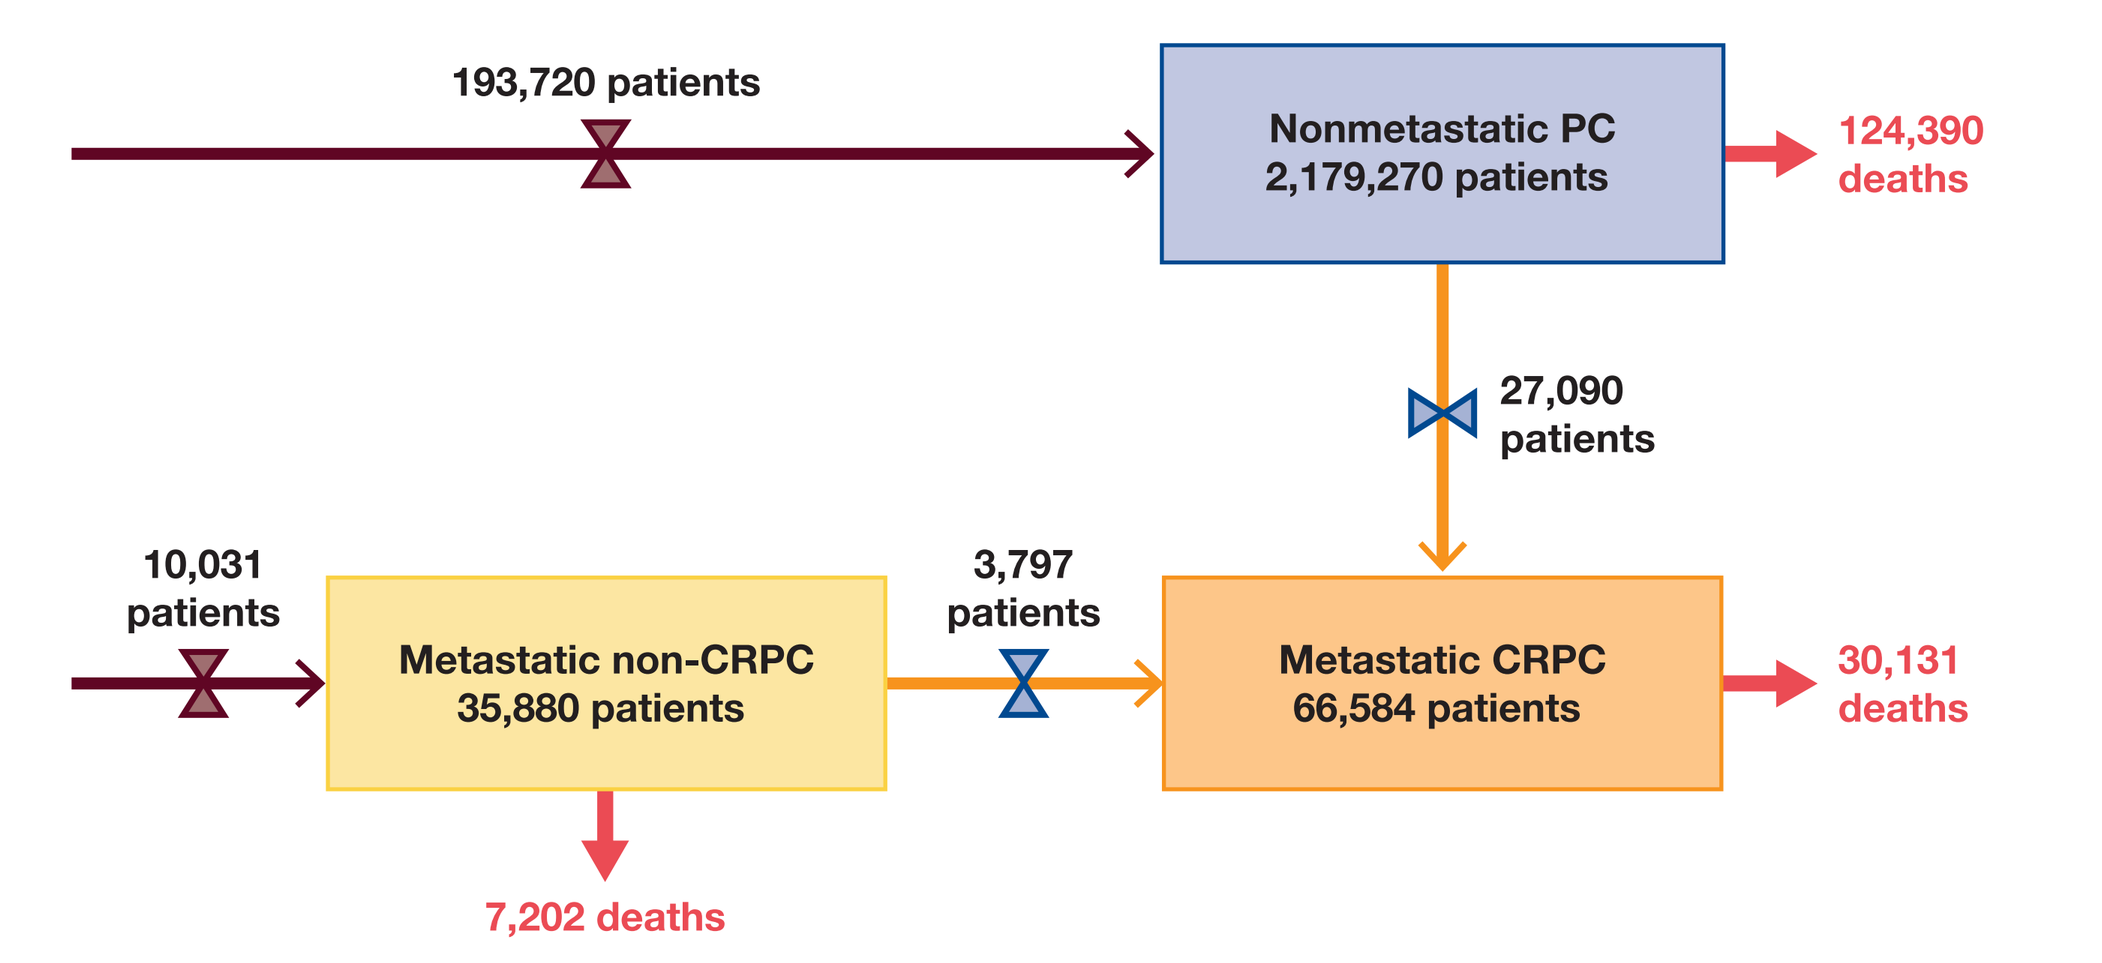
193,720 patients
Nonmetastatic PC
2,179,270 patients
124,390 deaths
27,090 patients
10,031 patients
Metastatic non-CRPC
35,880 patients
3,797 patients
Metastatic CRPC
66,584 patients
30,131 deaths
7,202 deaths

CONCLUSIONS

- This easily adaptable model can quantify the prevalence of distinct prostate cancer clinical states at any point in time, by accounting for patient movement through the disease continuum in a dynamic fashion. The results for overall prostate cancer prevalence and mortality derived from these simulations are consistent with epidemiological data[19] and previously published analyses of prostate cancer attributable mortality (14%-30%),[20,21] thus underscoring the validity of the model.
 - Almost 20% of annual prostate cancer deaths were estimated to be due to progressive castration-resistant disease, indicating a considerable unmet need for additional treatment options in this setting.
 - Accordingly, therapies designed to slow the disease trajectory and prevent progression to more advanced metastatic or symptomatic states can be regarded as analogous to cure. This could be particularly beneficial for men with early localized, rising prostate-specific antigen (PSA), or minimally metastatic disease states.
 - The model incorporates clinical data on available therapies for each clinical state, and can be continually updated to account for, or to estimate the potential impact of, new, more effective therapies on disease progression and mortality. Furthermore, while this model was calibrated to the epidemiology of prostate cancer in the US, it is flexible and can be tailored to other countries based on country-specific disease incidence trends, state distribution at diagnosis, and treatment patterns.
 - For instance, in the US only 5%-6% of annual incident patients presented with metastatic prostate cancer at the time of initial diagnosis over the time of the simulation. This relatively low incidence may be due to the widespread use of PSA-based early detection strategies.[22]
- This model suggests that each year a substantial number of prostate cancer patients in the US progress to advanced metastatic disease. These patients can suffer from significant morbidity and have a high mortality. For these men, new, more effective systemic therapies are needed to improve outcomes.
- The model was developed using published information available at the time the analysis was conducted. A limitation of the current model is its heterogeneity among the early states. Efforts are continuing to further refine the model, including more detailed estimates of outcomes with different local and systemic approaches, and the addition of data estimating physician practice patterns. The planned refinements will also include a specific focus on men with rising PSA after definitive local treatments.

REFERENCES

1. SEER. Available at http://www.seer.cancer.gov/statfacts/html/prost.html.
2. Klotz L. *Curr Oncol.* 2010;17(Suppl 2):S11-S17.
3. Capitanio U, et al. *Prostate.* 2010;70:1371-1378.
4. Scher HI, Heller G. *Urology.* 2000;55:523-527.
5. Shappley WV, et al. *J Clin Oncol.* 2009;27:4980-4985.
6. Moreira DM, et al. *J Urol.* 2009;182:2250-2255.
7. Merglen A, et al. *Arch Intern Med.* 2007;167:1944-1950.
8. Bolla M, et al. *Lancet.* 2005;366:572-578.
9. Antonarakis ES, et al. *J Clin Oncol.* 2007;25:4998-5008.
10. Ross RW, et al. *Cancer.* 2008;112:1247-1253.
11. Pilepich MV, et al. *Int J Radiat Oncol Biol Phys.* 2001;50:1243-1252.
12. Nelson JB, et al. *Cancer.* 2008;113:2478-2487.
13. Madan RA, at al. *Clin Cancer Res.* 2008;14:4526-4531.
14. Oudard S, at al. *BJU Int.* 2009;103:1641-1646.
15. Petrylak DP, et al. *N Engl J Med.* 2004;351:1513-1520.
16. Sternberg CN, et al. *J Clin Oncol.* 2009;27:5431-5438.
17. Choueiri TK, et al. *Cancer.* 2009;115:981-987.
18. Tannock IF, et al. *N Engl J Med.* 2004;351:1502-1512.
19. Jemal A, et al. *CA-Cancer J Clin.* 2009;59:225-249.
20. Cooperberg MR, et al. *J Natl Cancer Inst.* 2009;101:878-887.
21. Ketchandji M, et al. *J Am Geriatr Soc.* 2009;57:24-30.
22. Etzioni R. *Cancer Causes Control.* 2008;19:175-181.

Acknowledgments

The authors would like to thank Nicole Cossrow (Johnson & Johnson) for her assistance in developing the disease progression model. Development of this model was funded by Janssen Global Services, LLC. Writing assistance was provided by D. Wolf, MSc, of PAREXEL, and was funded by Janssen Global Services, LLC.

Poster presented at the 2011 ASCO Annual Meeting, June 3-7, 2011, Chicago, IL ◆ Supported by funding from Janssen Global Services, LLC

H2725 Arbtn ASCO ABI SB08.indd 1
6/3/11 1:05 PM